Long term benefit from Erlotinib treatment is independent of prognostic factors and therapeutic response

Schumann C., MD [1], Heigener D., MD[2], Dittrich I., MD[3], Abenhardt W., MD[4], Dederke B., MD[5], Duell T., MD[6], Matzdorff A., MD[7], Raack B.W., MD[8], Steins M., MD[9], Schneider C.P., MD[10]

[1] Universitätsklinikum Ulm Zentrum, [2] Krankenhaus Großhansdorf, [3] Lungenklinik Lostau, [4] Dres.Peter Bojko Wolfgang Abenhardt Daniel Bosse u.w. [5] Evang. Lungenklinik Berlin, [6] Asklepios Fachkliniken, [7] Caritasklinik St.Theresia, [8] Praxis Bernd Wolfgang Raack, [9] Thoraxklinik-Heidelberg, [10] Zentralklinik Bad Berka, Germany

Introduction

Erlotinib (Tarceva®) is an orally-active targeted inhibitor of the epidermal growth factor receptor. In the TRUST study, a single arm open-label phase IV trial assessing Erlotinib treatment in over 6,586 patients with advanced non small-cell lung cancer (NSCLC), the median progression-free survival was 3.17 months. 20% of the TRUST study patient population had a progression free survival at 12 months. Based on these findings we have initiated this retrospective study, performed in Germany, in order to analyze the profile of long term survivors, thus optimizing the selection of NSCLC patients which will benefit the most from Erlotinib treatment.

Aim

To analyze the profile of patients surviving longer than 12 months with advances NSCLC treated with Erlotinib.

Method

Questionnaires were retrospectively filled in by physicians treating NSCLC-patients in Germany with Tarceva for more than one year. Information of patients' demographics, pre-treatment general condition, potential risk factors, tumor histology, differentiation and stage, symptoms, accompanying diseases, as well as details of the clinical examination, the basic laboratory tests, and imaging studies, were collected. Details of the patient's previous chemotherapy and reasons for its discontinuation were also collected. Finally, data related to Erlotinib treatment were evaluated: line of therapy, dosage, duration of therapy, type and duration of therapeutic response, symptoms and general condition before therapy and during the course of treatment, tolerability of the therapy, and current therapy status. The overall assessment from both patients and physicians perspectives were also noted.

Results

Data from 301 patients were collected.

The average age of patients surviving over 12 months was 66 years (range 23-87). 75% of patients were in good or moderately restricted conditions (Eastern Cooperative Oncology Group score 0 or 1). The treatment duration shows, that 52% had been treated with Erlotinib for at least 18 months, and 25% were treated for over 24 months; 50% of the patients received Erlotinib as second line therapy.

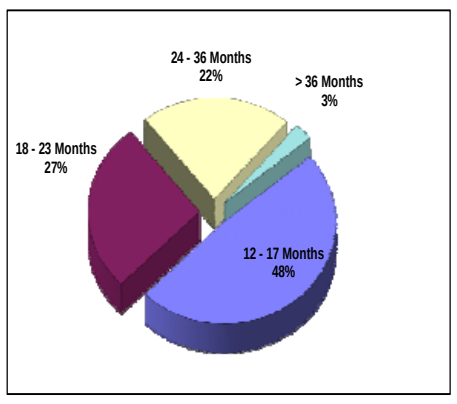

24 - 36 Months 22%
> 36 Months 3%
18 - 23 Months 27%
12 - 17 Months 48%

Therapy duration under Erlotinib treatment

The long-term survival with Erlotinib was independent of the prognostic factors of gender, tumor histology and smoking status. 43% of patients were male, 14% were current smokers and 34% were past smokers. Histology type was adenocarcinoma and squamous cell in 67% and 16% of patients, respectively.

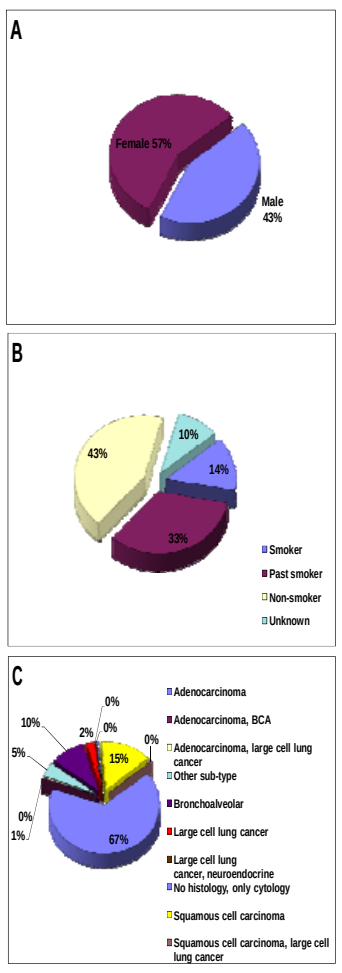

A
Female 57%
Male 43%
B
43%
10%
14%
33%
Smoker
Past smoker
Non-smoker
Unknown
C
Adenocarcinoma
Adenocarcinoma, BCA
Adenocarcinoma, large cell lung cancer
Other sub-type
Bronchoalveolar
Large cell lung cancer
Large cell lung cancer, neuroendocrine
No histology, only cytology
Squamous cell carcinoma
Squamous cell carcinoma, large cell lung cancer
67%
15%
10%
5%
2%
1%
0%

Gender (A), smoking status (B) and tumor histology (C) in patients surviving over 12 months under Erlotinib treatment

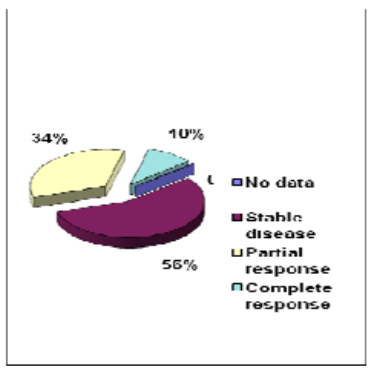

34%
10%
56%
No data
Stable disease
Partial response
Complete response

Therapeutic response in patients surviving over 12 months

Long-term survival was also independent of the therapeutic response. 56% of patients had a stable disease over the course of treatment, while 44% had partial or full response.

Although 78% of patients developed a typical skin reaction, the rash was well controlled even in severe cases, and the tolerability of Erlotinib was considered good or very good by 77% of treating physicians.

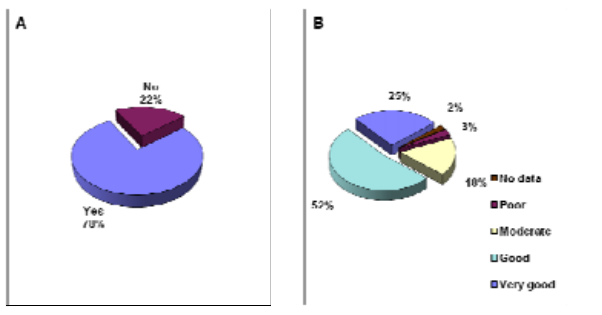

A
No 22%
B
25%
2%
3%
18%
52%
No data
Poor
Moderate
Good
Very good

Development of rash (A) and tolerability (B) to Erlotinib treatment

Conclusion

This study analyzed the characteristics of patients who were treated for at least one year with Erlotinib, in order to better characterize the patient groups which will benefit most of this target-oriented therapy.

According to the results of this analysis, the long-term benefit of Erlotinib, which was predominantly very well or well tolerated, was not limited to groups with good prognostic factors. Long-term survivors also included patients with unfavorable risk factors. The relatively high proportion of males (42%), smokers or past-smokers (45%) and patients with squamous cell carcinoma (15%) is remarkable, because, to date, Erlotinib is offered less often to these patients in daily practice. This is indicative of the broad efficacy of Erlotinib across all patients subgroups.

The long-term benefit of Erlotinib, according to the results of the present analysis, is independent of the type of therapeutic response. By contrast to conventional chemotherapy, long-term therapeutic success under Erlotinib was not restricted to patients with a complete or partial remission.

Although most of the patients treated with Erlotinib develop cutaneous side effects with variable intensity, this study confirms that these skin reactions are well controllable and tolerable in day-to-day practice, even for treatment durations of more than one year.

In summary, these data confirm Erlotinib to be effective in the long term treatment of NSCLC.